Impact of Bulevirtide Given With or Without Nucleos(t)ide Analogues on 48-Week Virologic Outcomes in Patients With Chronic Hepatitis Delta Virus Infection

TOP-400

Pietro Lampertico[1,2], Maurizia Brunetto[3,4], Maria Buti[5,6], Soo Aleman[7], Pavel Bogomolov[8], Vladimir Chulanov[9], Nina Mamonova[10], Viacheslav Morozov[11], Olga Sagalova[12], Tatyana Stepanova[13], John F Flaherty[14], Mingyang Li[14], Dmitry Manuilov[14], Ben L Da[14], Renee-Claude Mercier[14], Grace M Chee[14], Markus Cornberg[15], Heiner Wedemeyer[15], Tarik Asselah[16]

[1]Division of Gastroenterology and Hepatology, Foundation IRCCS Ca' Granda Ospedale Maggiore Policlinico, Milan, Italy; [2]Department of Pathophysiology and Transplantation, CRC "A. M. and A. Migliavacca" Center for Liver Disease, University of Milan, Milan, Italy; [3]Hepatology Unit, Reference Center of the Tuscany Region for Chronic Liver Disease and Cancer, University Hospital of Pisa, Pisa, Italy; [4]Department of Clinical and Experimental Medicine, University of Pisa, Pisa, Italy; [5]Hospital Universitario Vall d'Hebron, Barcelona, Spain; [6]CIBEREHD del Instituto Carlos III, Madrid, Spain; [7]Department of Infectious Diseases, Karolinska University Hospital/Karolinska Institutet, Stockholm, Sweden; [8]M.F. Vladimirsky Moscow Regional Research and Clinical Institute, Moscow, Russian Federation; [9]Sechenov University, Moscow, Russian Federation; [10]FSBI National Research Medical Center for Phthisiopulmonology and Infectious Diseases of the Ministry of Health of the Russian Federation, Moscow, Russian Federation; [11]LLC Medical Company "Hepatolog," Samara, Russian Federation; [12]South Ural State Medical University, Chelyabinsk, Russian Federation; [13]LLC Clinic of Modern Medicine, Moscow, Russian Federation; [14]Gilead Sciences, Inc., Foster City, CA, USA; [15]Medizinische Hochschule Hannover, Klinik für Gastroenterologie, Hepatologie und Endokrinologie, Hannover, Germany; [16]Hôpital Beaujon APHP, Université de Paris-Cité, INSERM, Clichy, France

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

Conclusions

- Concomitant NA therapy does not impact HDV responses with BLV treatment but may lead to greater reductions in HBV DNA levels and higher rates of undetectable HBV DNA and HBV DNA <LLOQ
- BLV treatment with or without NA therapy had no appreciable impact on HBsAg levels

Plain Language Summary

- Patients with chronic hepatitis delta virus received bulevirtide 2 or 10 mg or no anti–hepatitis delta virus treatment and were grouped by those who did or did not also receive nucleos(t)ide analogue therapy
- Receiving nucleos(t)ide analogue therapy with bulevirtide treatment did not have a noticeable impact on hepatitis delta virus responses
- At week 48, levels of hepatitis B virus DNA declined in patients who received bulevirtide with or without nucleos(t)ide analogue therapy
- Greater declines in hepatitis B virus DNA levels and higher rates of hepatitis B viral suppression occurred at week 48 in patients treated with both bulevirtide and nucleos(t)ide analogue therapy
- Levels of hepatitis B surface antigen in patients were mostly unimpacted by bulevirtide treatment with or without nucleos(t)ide analogue therapy

References: **1.** Stockdale AJ, et al. *J Hepatol.* 2020;73:523-32. **2.** Wedemeyer H, et al. *N Engl J Med.* 2024;389:22-32. **3.** Hepcludex (bulevirtide). European Medicines Agency. Gilead Sciences, Inc.; 2023. **4.** Terrault N, et al. *Hepatology.* 2018;67(4):1560-99. **5.** European Association for the Study of the Liver. *J Hepatol.* 2023;79:433-60.

Acknowledgments: We extend our thanks to the patients, their families, and all participating investigators and their corresponding site staff. Writing and editorial support were provided by Molly Yeager, PhD, and Danielle Shepherd, PhD, of AlphaScientia, a Red Nucleus company, and were funded by Gilead Sciences, Inc. This study was funded by Gilead Sciences, Inc.

Disclosures: **PL** reports speaking and teaching fees from and participation in advisory committees or review panels for AbbVie; Aligos Therapeutics; Alnylam Pharmaceuticals; Antios Therapeutics; Arrowhead Pharmaceuticals; Bristol Myers Squibb; Eiger Biopharmaceuticals; Gilead Sciences, Inc.; GSK; Janssen; Merck Sharp & Dohme; MYR GmbH; Roche; and Spring Bank Pharmaceuticals. **MBr** reports consulting and serving on a speakers bureau for AbbVie; Eisai-Merck Sharp & Dohme; Gilead Sciences, Inc.; Janssen; and Roche. **MBu** reports speaker fees, research support, and consulting fees from AbbVie; Gilead Sciences, Inc.; and Janssen. **SA** reports speaking honoraria from AbbVie; Biogen; Gilead Sciences, Inc.; and Merck Sharp & Dohme, and research grants from AbbVie and Gilead Sciences, Inc. **PB** reports nothing to disclose. **VC** reports being a consultant and giving sponsored lectures for AbbVie; AstraZeneca; Bristol Myers Squibb; Gilead Sciences, Inc.; GSK; Hepatera; Merck Sharp & Dohme; Roche; and R-Pharm. **NM**, **VM**, **OS**, and **TS** report nothing to disclose. **JFF**, **ML**, **DM**, **BLD**, **R-CM**, and **GMC** are employees of Gilead Sciences, Inc., and may own stock in Gilead Sciences, Inc. **MC** reports honoraria from AbbVie; Falk; Gilead Sciences, Inc.; GSK; Janssen-Cilag; Merck Sharp & Dohme; Novartis; Roche; Spring Bank Pharmaceuticals; and Swedish Orphan Biovitrum. **HW** reports honoraria for speaking or consulting from Abbott; AbbVie; Boehringer Ingelheim; Bristol Myers Squibb; Eiger Biopharmaceuticals; Gilead Sciences, Inc.; Janssen; Merck Sharp & Dohme; MYR GmbH; Novartis; Novira; Roche; Roche Diagnostics; Siemens; and Transgene; and research support from Abbott; Bristol Myers Squibb; and Gilead Sciences, Inc. **TA** reports acting as a speaker and investigator for AbbVie; Eiger Biopharmaceuticals; Gilead Sciences, Inc.; Janssen; Merck; MYR Pharmaceuticals; and Roche.

Introduction

- Hepatitis delta virus (HDV) represents the most severe form of chronic viral hepatitis and is estimated to affect between 10 and 20 million individuals worldwide[1]
- In the Phase 3 Study MYR301, patients with chronic hepatitis delta (CHD) randomised to receive bulevirtide (BLV; 2 or 10 mg), a novel entry inhibitor of HDV given once daily subcutaneously, had a superior treatment response compared with the delayed-treatment (control) group by the primary endpoint of combined response at week (W) 48 (defined as HDV RNA decline ≥2 $\log_{10}$ IU/mL from baseline [BL] or undetectable HDV RNA with alanine aminotransferase [ALT] normalisation)[2]
- BLV is now fully approved in the EU, Great Britain, Switzerland, and the Russian Federation at the dose of 2 mg/d for the treatment of CHD with compensated liver disease[3]
- Patients with CHD may receive concomitant nucleos(t)ide analogues (NAs), such as tenofovir disoproxil fumarate (TDF), tenofovir alafenamide (TAF), or entecavir, to treat the underlying hepatitis B virus (HBV) infection, but while these are considered first-line therapies for chronic hepatitis B,[4] they generally have not demonstrated any effect on HDV responses[5]

Objective

- This study aims to characterise the impact of NA therapy on HBV and HDV responses in patients with CHD treated with BLV

Methods

- A pooled analysis of the following 3 clinical trials was performed: MYR203 (NCT02888106), MYR204 (NCT03852433), and MYR301 (NCT03852719)
- Data from patients with CHD treated with BLV 2 or 10 mg monotherapy or who received no anti-HDV treatment (MYR301 control arm) were included
- In MYR204 and MYR301, concomitant use of NA therapy was allowed at the discretion of the investigator based on guidelines for HBV treatment, and in MYR203, TDF was administered based on study arm
- HDV RNA levels were determined by RT-qPCR using RoboGene® HDV RNA Quantification Kit 2.0; HBV DNA levels were determined by the Abbott RealTime HBV Viral Load Assay on an Abbott m2000 RealTime System (Abbott Diagnostics); hepatitis B surface antigen (HBsAg) levels were determined using the ARCHITECT HBsAg Next Qualitative assay (Abbott Diagnostics)
- The primary analysis focused on the impact of NA therapy on the following:
 - — HDV outcomes, including:
 - ○ Virologic response (VR; undetectable HDV RNA or a ≥2 $\log_{10}$ IU/mL decline in HDV RNA from BL)
 - ▪ Undetectable HDV RNA: <lower limit of quantification (LLOQ; 50 IU/mL) with target not detected in the MYR204 and MYR301 studies, or below the limit of detection (LOD; 10 IU/mL) in the MYR203 study
 - ○ ALT normalisation
 - ▪ ALT ≤31 U/L for females and ≤41 U/L for males at Russian sites and ≤34 U/L for females and ≤49 U/L for males at all other sites
 - ○ Combined response (defined as VR with ALT normalisation)
 - — HBV outcomes, including:
 - ○ HBV DNA
 - ▪ Undetectable HBV DNA: <LLOQ (10 IU/mL) with target not detected
 - ○ Change in HBV DNA and HBsAg levels from BL
 - ▪ Change in HBV DNA from BL was evaluated only in patients with HBV DNA ≥LLOQ at BL

Results

- Overall, 280 patients were included across 3 groups for this integrated analysis:
 - — BLV treated (n = 229): BLV 2 mg/d (n = 64), BLV 10 mg/d (n = 165)
 - — No anti-HDV treatment (control, n = 51)
- Among all patients,160 (57%; range across study groups, 48% to 63%) received concomitant NA therapy
 - — Of these, 87% (139/160) received a TDF/TAF-containing NA therapy

Demographics and Baseline Characteristics (BLV Treated and Control)

	BLV 2 mg (n = 64)	BLV 10 mg (n = 165)	Control (n = 51)	BLV Treated (2 + 10 mg) (n = 229)	Total Cohort (N = 280)
Age, years, mean (SD)	43 (9.1)	40 (8.3)	41 (7.5)	41 (8.6)	41 (8.4)
Male sex, n (%)	41 (64.1)	105 (63.6)	26 (51.0)	146 (63.8)	172 (61.4)
Race, n (%)					
White	56 (87.5)	141 (85.5)	40 (78.4)	197 (86.0)	237 (84.6)
Asian	8 (12.5)	21 (12.7)	11 (21.6)	29 (12.7)	40 (14.3)
Black or African American	0	3 (1.8)	0	3 (1.3)	3 (1.1)
Cirrhosis present,[a] n (%)	26 (40.6)	65 (39.4)	24 (47.1)	91 (39.7)	115 (41.1)
HBeAg positive, n (%)	7 (10.9)	18 (10.9)	4 (7.8)	25 (10.9)	29 (10.4)
Concomitant NA therapy, n (%)	31 (48.4)	97 (58.8)	32 (62.7)	128 (55.9)	160 (57.1)
Prior IFN therapy, n (%)	27 (42.2)	79 (47.9)	29 (56.9)	106 (46.3)	135 (48.2)
Genotype HDV-1,[b] n (%)	64 (100.0)	159 (96.4)	51 (100.0)	223 (97.4)	274 (97.9)
HBV DNA, $\log_{10}$ IU/mL, mean (SD)	1.4 (1.3)	1.3 (1.4)	0.9 (1.0)	1.3 (1.4)	1.2 (1.3)
HDV RNA, $\log_{10}$ IU/mL, mean (SD)	5.2 (1.3)	5.2 (1.5)	5.1 (1.4)	5.2 (1.4)	5.2 (1.4)
HBsAg, $\log_{10}$ IU/mL, mean (SD)	3.8 (0.5)	3.7 (0.7)	3.7 (0.5)	3.7 (0.6)	3.7 (0.6)
ALT, U/L, median (Q1, Q3)	86 (66, 137)	83 (57, 119)	80 (57, 116)	84 (61, 127)	83 (61, 125)
LSM, kPa, mean (SD)	14.0 (7.8)	14.1 (9.3)	15.3 (9.0)	14.0 (8.9)	14.3 (8.9)

[a]Cirrhosis status was missing for 15 patients in the BLV 10 mg group as data on cirrhosis status were not collected for patients who received BLV 5 mg BID + TDF 300 mg QD in the MYR203 study. [b]BLV 10 mg group: HDV-5, n = 2; missing, n = 4.
ALT, alanine aminotransferase; BID, twice daily; BLV, bulevirtide; HBeAg, hepatitis B e antigen; HBsAg, hepatitis B surface antigen; HBV, hepatitis B virus; HDV, hepatitis delta virus; IFN, interferon; LSM, liver stiffness measurement; NA, nucleos(t)ide analogue; Q, quartile; QD, once daily; TDF, tenofovir disoproxil fumarate.

- Patient demographics and BL characteristics were relatively well balanced across treatment groups

Demographics and Baseline Characteristics by Concomitant NA Therapy (BLV Treated)

	Concomitant NA Therapy (n = 128)	No Concomitant NA Therapy (n = 101)
Age, years, mean (SD)	41 (9.2)	41 (7.8)
Male sex, n (%)	82 (64.1)	64 (63.4)
Race, n (%)		
White	104 (81.3)	93 (92.1)
Asian	21 (16.4)	8 (7.9)
Black or African American	3 (2.3)	0
Cirrhosis present,[a] n (%)	62 (48.4)	29 (28.7)
HBeAg positive, n (%)	20 (15.6)	5 (5.0)
Prior IFN therapy, n (%)	69 (53.9)	37 (36.6)
Genotype HDV-1,[b] n (%)	122 (95.3)	101 (100.0)
HBV DNA, $\log_{10}$ IU/mL, mean (SD)	1.2 (1.5)	1.4 (1.1)
HDV RNA, $\log_{10}$ IU/mL, mean (SD)	5.2 (1.4)	5.2 (1.4)
HBsAg, $\log_{10}$ IU/mL, mean (SD)	3.7 (0.6)	3.7 (0.7)
ALT, U/L, median (Q1, Q3)	88 (60, 126)	83 (63, 127)
LSM, kPa, mean (SD)	14.5 (9.5)	13.4 (7.9)

[a]Cirrhosis status was missing for 15 patients with concomitant NA therapy as data on cirrhosis status were not collected for patients who received BLV 5 mg BID + TDF 300 mg QD in the MYR203 study.
[b]Concomitant NA therapy group: HDV-5, n = 2; missing, n = 4.
ALT, alanine aminotransferase; BID, twice daily; BLV, bulevirtide; HBeAg, hepatitis B e antigen; HBsAg, hepatitis B surface antigen; HBV, hepatitis B virus; HDV, hepatitis delta virus; IFN, interferon; LSM, liver stiffness measurement; NA, nucleos(t)ide analogue; Q, quartile; QD, once daily; TDF, tenofovir disoproxil fumarate.

- Patients who received concomitant NA therapy were more likely to be Asian, cirrhotic, and HBeAg positive or to have a prior history of IFN therapy

HDV Responses at W48 by NA Therapy (BLV Treated)

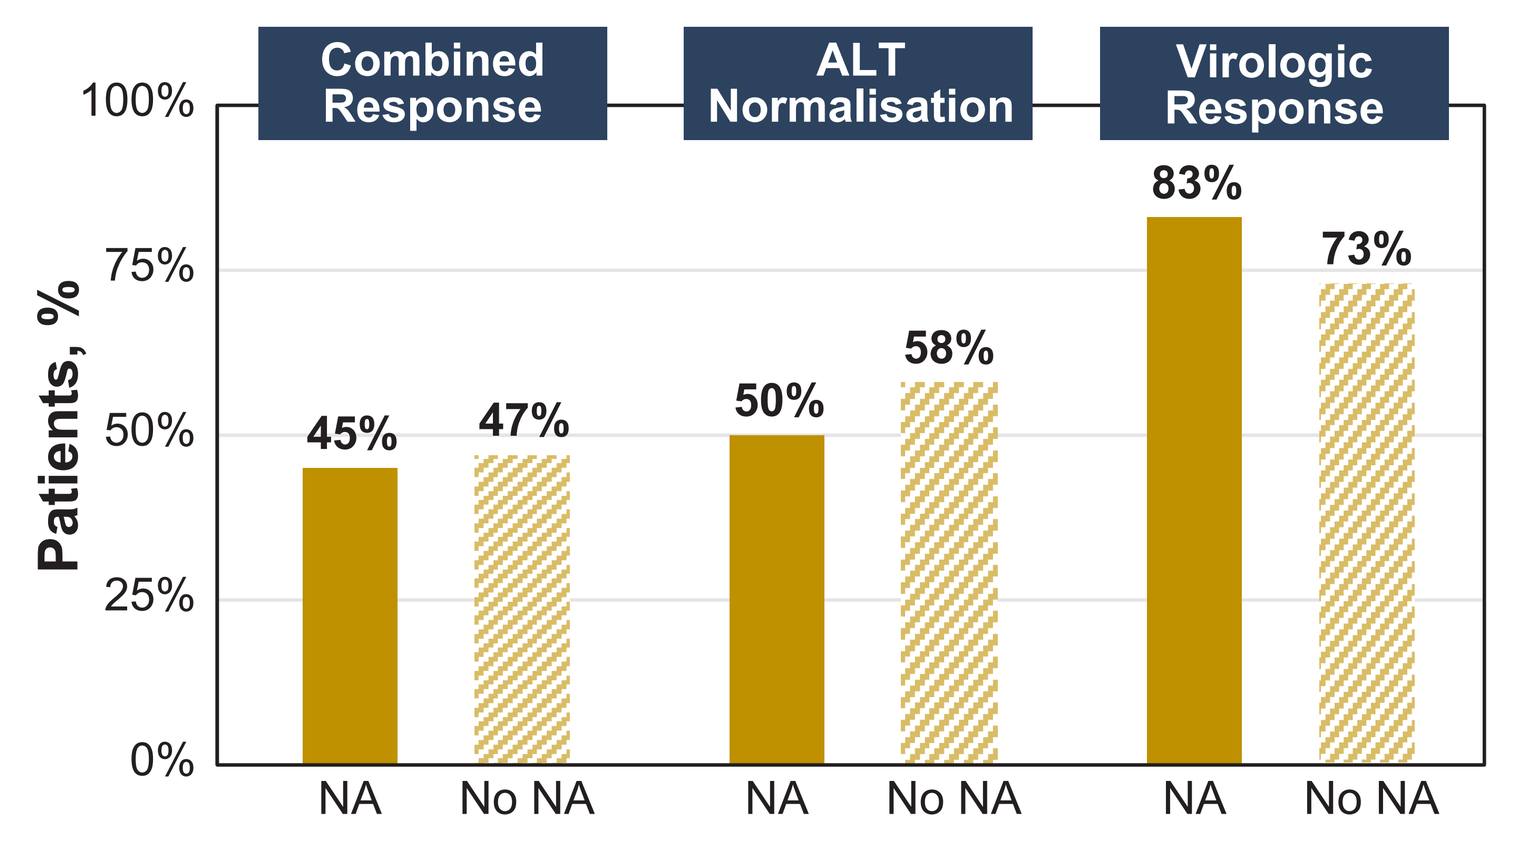
Combined Response
ALT Normalisation
Virologic Response
100%
75%
50%
25%
0%
Patients, %
45%
47%
50%
58%
83%
73%
NA
No NA
NA
No NA
NA
No NA

NA, n = 128. No NA, n = 101. Missing data were considered nonresponders.
ALT, alanine aminotransferase; BLV, bulevirtide; HDV, hepatitis delta virus; NA, nucleos(t)ide analogue; W, week.

HDV Responses at W48 by NA Therapy (Control)

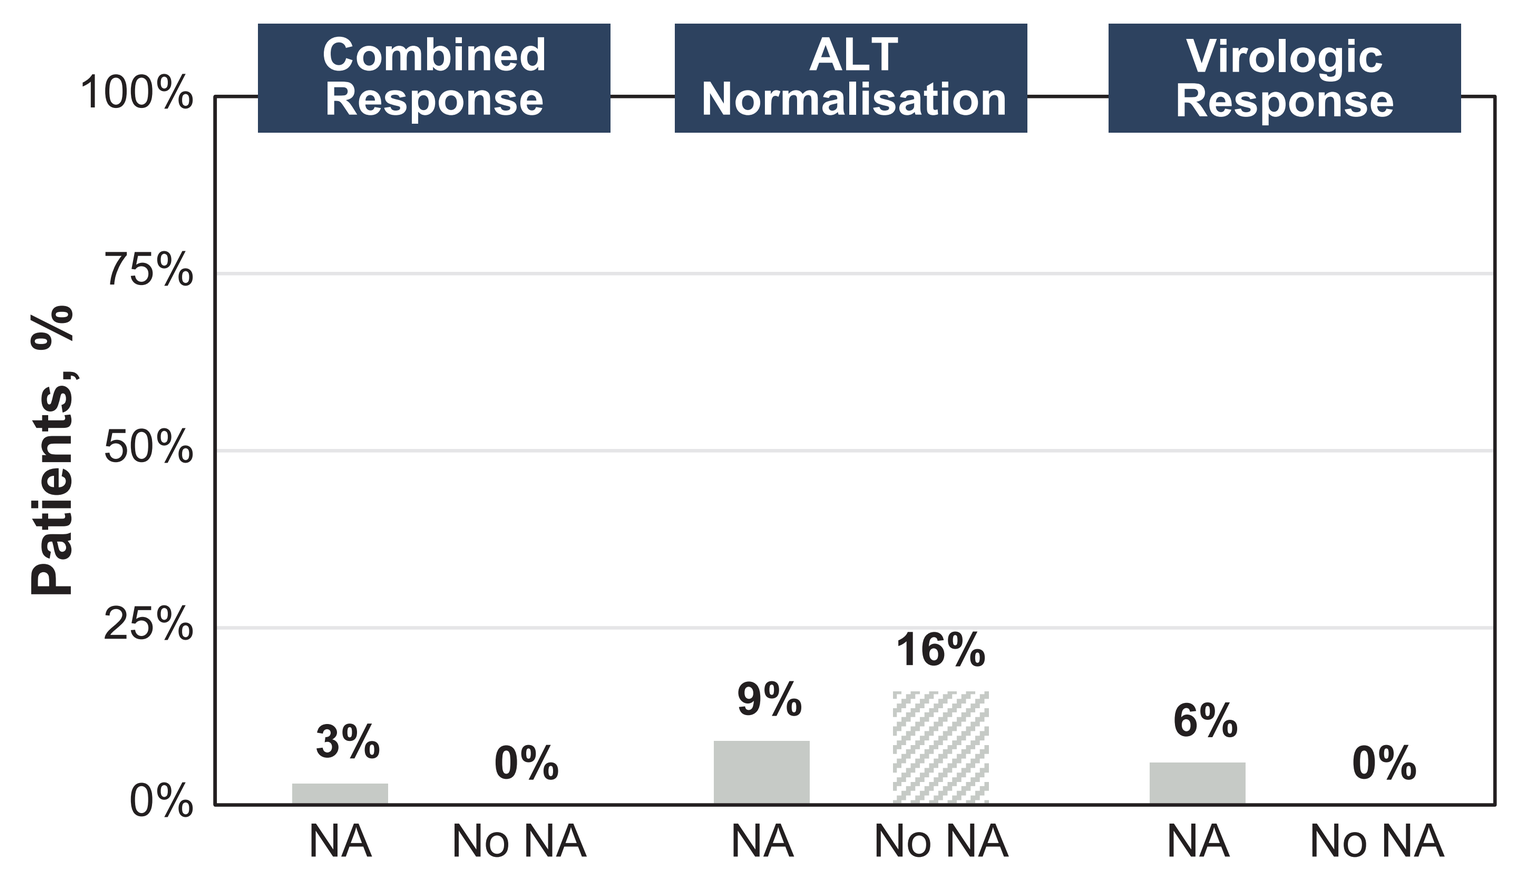
Combined Response
ALT Normalisation
Virologic Response
100%
75%
50%
25%
0%
Patients, %
3%
0%
9%
16%
6%
0%
NA
No NA
NA
No NA
NA
No NA

NA, n = 32. No NA, n = 19. Missing data were considered nonresponders.
ALT, alanine aminotransferase; HDV, hepatitis delta virus; NA, nucleos(t)ide analogue; W, week.

- Concomitant NA therapy in both BLV-treated patients and the control group did not have an appreciable impact on HDV responses

Mean HBV DNA Change From BL at W48 by NA Therapy[a]

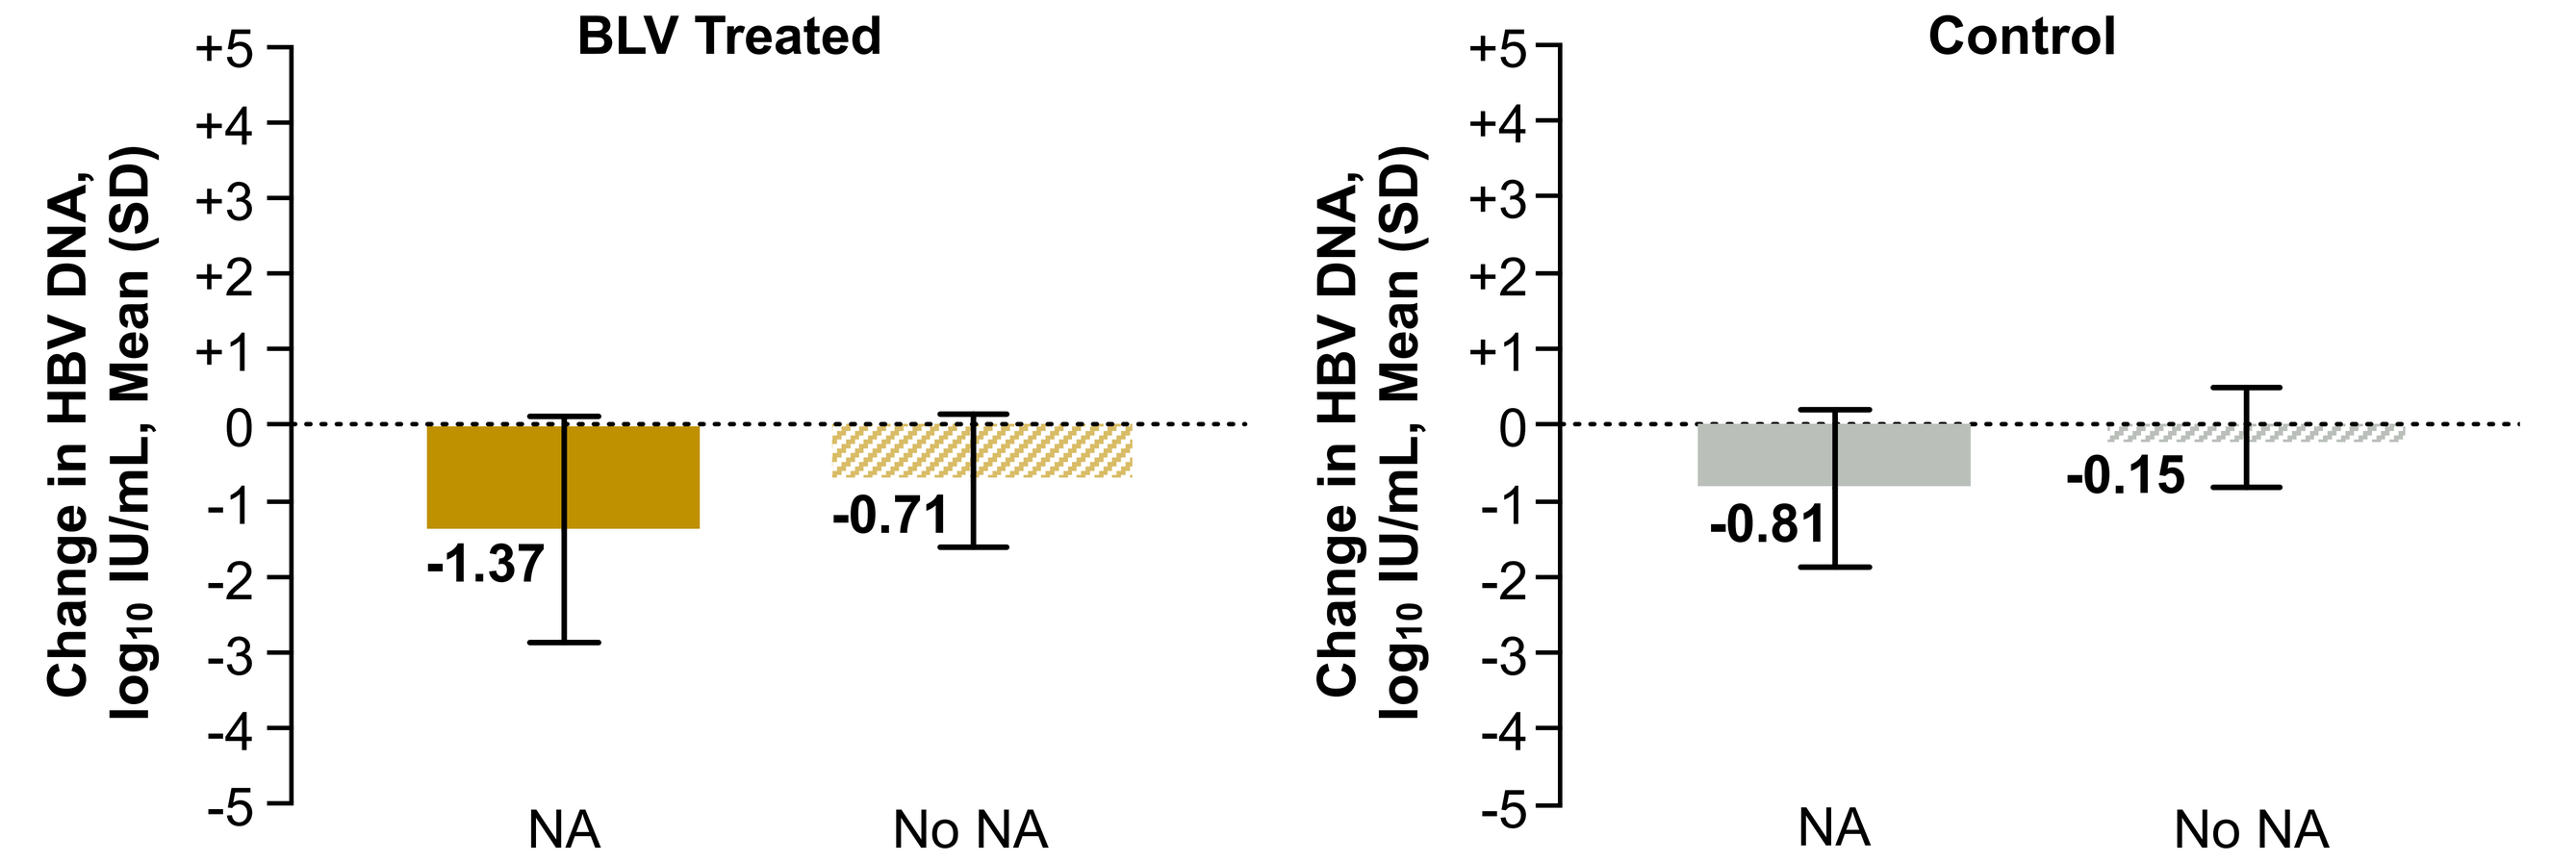
BLV Treated
Control
Change in HBV DNA, $\log_{10}$ IU/mL, Mean (SD)
+5
+4
+3
+2
+1
0
-1
-2
-3
-4
-5
-1.37
-0.71
-0.81
-0.15
NA
No NA
NA
No NA

BLV treated: NA, n = 73; no NA, n = 72. Control: NA, n = 15; no NA, n = 12.
[a]Change in HBV DNA from BL was evaluated only in patients with HBV DNA ≥LLOQ at BL.
BL, baseline; BLV, bulevirtide; HBV, hepatitis B virus; LLOQ, lower limit of quantification; NA, nucleos(t)ide analogue; W, week.

- HBV DNA levels declined at W48 in patients treated with BLV with or without NAs, with greater declines observed in patients treated with BLV + NA therapy

HBV Responses at W48 by NA Therapy (BLV Treated)

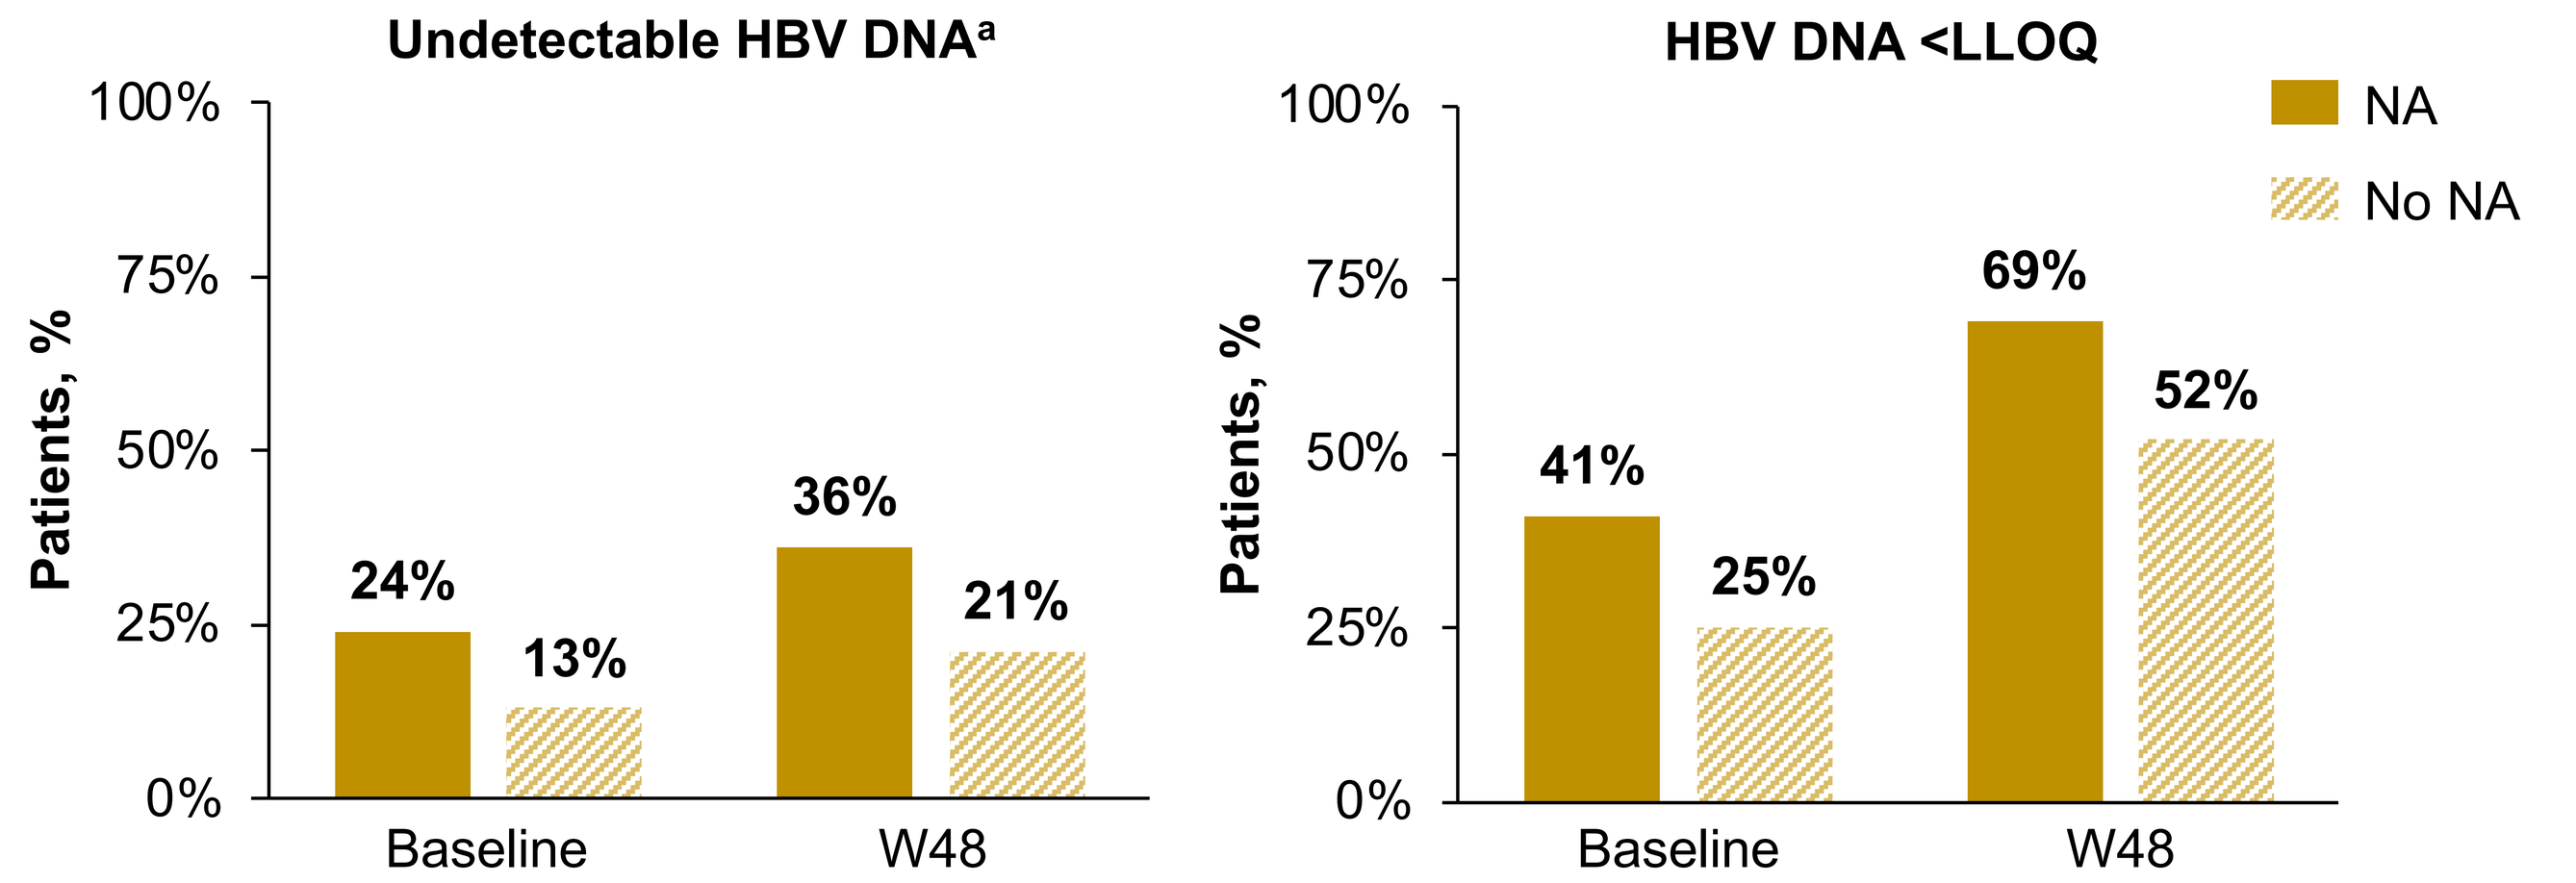
Undetectable HBV DNA[a]
HBV DNA <LLOQ
NA
No NA
100%
75%
50%
25%
0%
Patients, %
24%
13%
36%
21%
41%
25%
69%
52%
Baseline
W48
Baseline
W48

NA: baseline, n = 126; W48: n = 124. No NA: baseline, n = 100; W48, n = 97.
One patient's baseline time point was evaluated using an assay with a LLOQ of 100 IU/mL; in this case, the target was detected and the patient was classified appropriately.
[a]Undetectable HBV DNA: <LLOQ (10 IU/mL) with target not detected.
BLV, bulevirtide; HBV, hepatitis B virus; LLOQ, lower limit of quantification; LOD, limit of detection; NA, nucleos(t)ide analogue; W, week.

- Increasing rates of undetectable HBV DNA and HBV DNA <LLOQ from BL to W48 were observed in patients treated with BLV with or without NAs; the highest rates were observed in patients treated with BLV + NA therapy

HBV Responses at W48 by NA Therapy (Control)

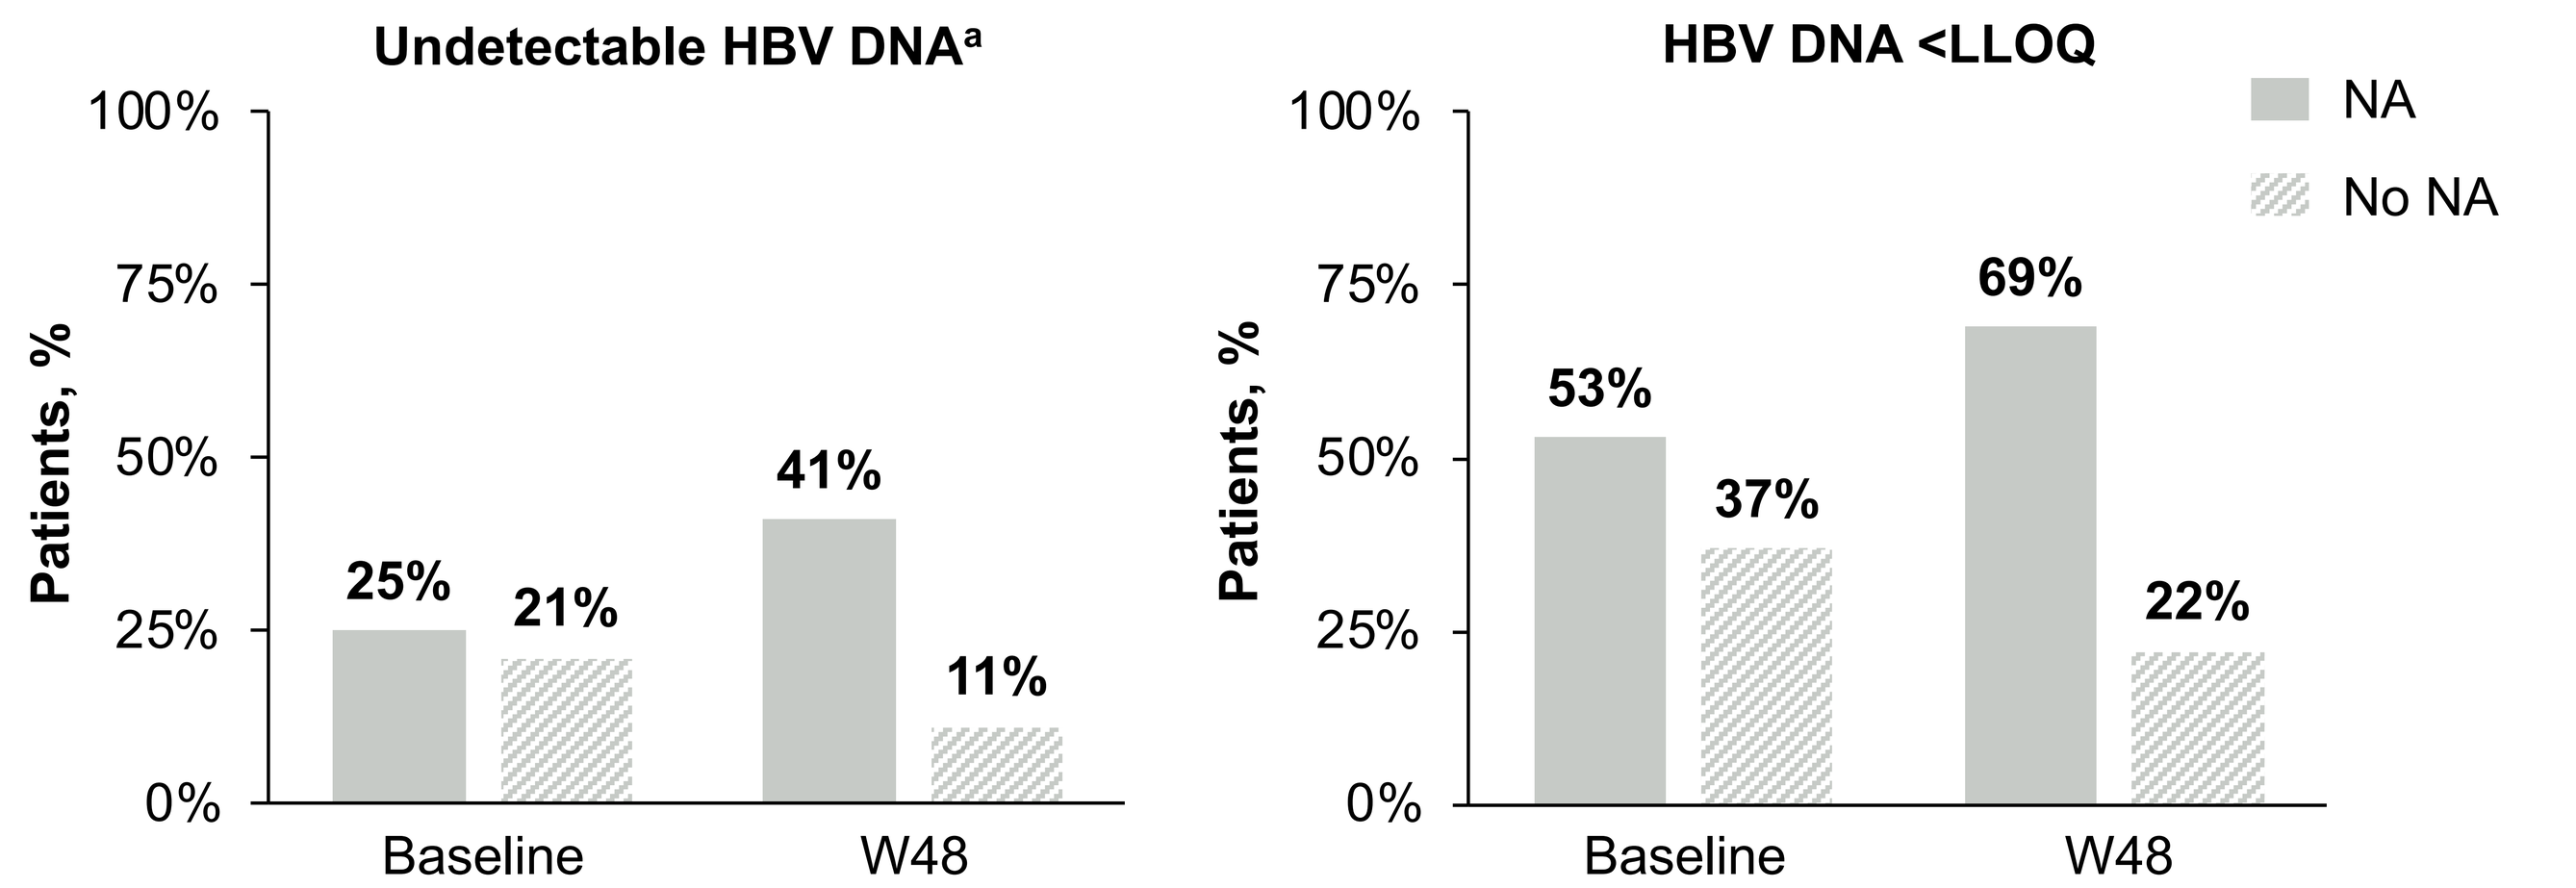
Undetectable HBV DNA[a]
HBV DNA <LLOQ
NA
No NA
100%
75%
50%
25%
0%
Patients, %
25%
21%
41%
11%
53%
37%
69%
22%
Baseline
W48
Baseline
W48

NA: baseline and W48, n = 32. No NA: baseline, n = 19; W48, n = 18.
[a]Undetectable HBV DNA: <LLOQ (10 IU/mL) with target not detected.
HBV, hepatitis B virus; LLOQ, lower limit of quantification; NA, nucleos(t)ide analogue; W, week.

- Rates of undetectable HBV DNA and HBV DNA <LLOQ declined over time in control patients who did not receive BLV or NAs

Mean HBsAg Change From BL at W48 by NA Therapy

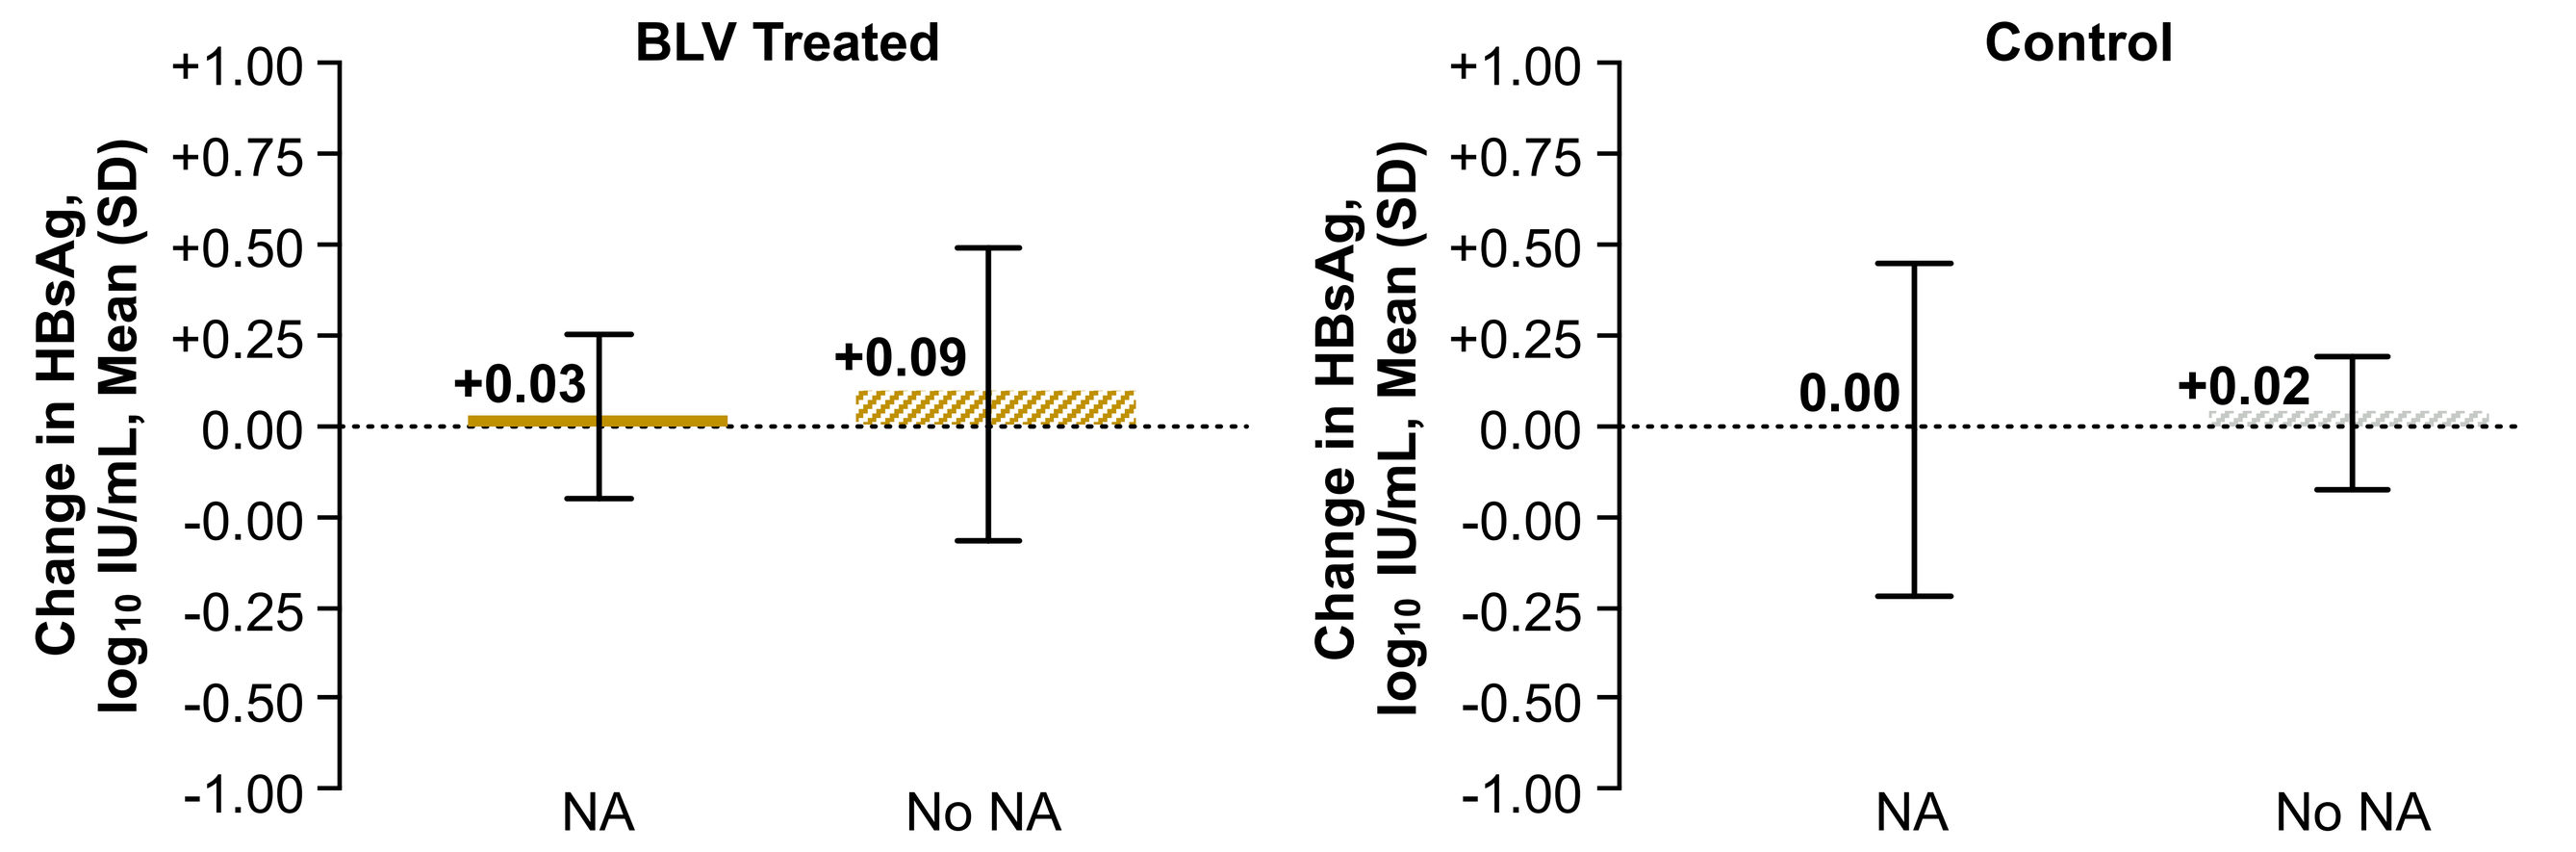
BLV Treated
Control
Change in HBsAg, $\log_{10}$ IU/mL, Mean (SD)
+1.00
+0.75
+0.50
+0.25
0.00
-0.00
-0.25
-0.50
-1.00
+0.03
+0.09
0.00
+0.02
NA
No NA
NA
No NA

BLV treated: NA, n = 121; no NA, n = 95. Control: NA, n = 32; no NA, n = 18.
BL, baseline; BLV, bulevirtide; HBsAg, hepatitis B surface antigen; NA, nucleos(t)ide analogue; W, week.

- BLV monotherapy with or without NA therapy had no appreciable impact on HBsAg levels

European Association for the Study of the Liver; 05–08 June 2024; Milan, Italy